Cost-effectiveness analysis of empagliflozin in comparison to liraglutide based on cardiovascular outcome trials

PDB76

Mark Lamotte[1], Mafalda Ramos[1], Ahmed Salem[1], Volker Foos[1], Anastasia Ustyugova[2], Nikco Hau[3], Pranav Gandhi[4]

[1]IQVIA Real World Evidence Solutions, Zaventem, Belgium; [2]Boehringer Ingelheim International GmbH, Ingelheim, Germany; [3]Boehringer Ingelheim Ltd, Bracknell, UK; [4]Boehringer Ingelheim Pharmaceuticals Inc., Ridgefield, CT, USA.

Background

- Management of type 2 diabetes (T2D) is balanced between achieving and maintaining target glycaemic control (haemoglobin A1c (HbA1c)) to avoid or delay the onset of costly macro- and microvascular complications, and avoiding hypoglycaemia. For new treatments addressing both components is critical in T2D management.
- Over the recent years, several cardiovascular outcome trials (CVOTs) have been published with glucose lowering drugs.
- In the EMPA-REG OUTCOME trial[1,2], empagliflozin (SGLT2) + standard of care (SoC) was compared to SoC (placebo arm in the trial) in patients with type 2 diabetes (T2D) and established cardiovascular disease (CVD).
- The LEADER trial[3] for liraglutide (GLP1) was designed similarly i.e. in patients with T2D and established CVD.

Aim

To assess the short-term (5 years) cost-effectiveness of empagliflozin + standard of care (SoC) in adult patients with T2D and established CVD compared to liraglutide + SoC from the UK NHS perspective

Method

CDM overview

- The IQVIA Core Diabetes Model (CDM) CVO v9.0, a well-established microsimulation model with 17 interdependent Markov sub-models was used to capture major complications (CV, renal, ophthalmological, nephropathy etc.) of diabetes along with management cost and results were generated for incremental costs, life expectancy (LE), and quality-adjusted life years (QALYs)
- The model was calibrated to reproduce the 3-year event rates (recalculated from the event rates per 1000 patient years) from the EMPA-REG OUTCOME trial, for empagliflozin + SoC.
- A Network Meta-Analysis (NMA) provided the relative risks for CVD outcomes with empagliflozin versus liraglutide[4]. The CDM was then calibrated to reproduce liraglutide trials' outcomes.
- In Table 1 the results of the calibration are shown.

Table 1. CDM predicted 3-year event rates compared to original EMPA-REG OUTCOME and indirect comparison results (%)

	EMPAGLIFLOZIN		LIRAGLUTIDE	
	EMPA-REG OUTCOME observed	Model predicted	Estimated by NMA	Model predicted
Death from any cause	5.82	5.78	7.28	7.24
Death from CV cause	3.72	3.68	4.65	4.63
Myocardial Infarction	5.04	5.05	5.09	5.08
Hospitalization for Unstable Angina	3.00	3.01	3.00	3.06
Stroke	3.69	3.70	2.72	2.71
Hospitalization for Heart Failure	2.82	2.83	3.76	3.79
Microalbuminuria	75.75	75.86	75.75	76.76
End stage renal disease	0.3	0.3	0.3	0.28

Analytic Overview

- The UK NHS perspective was taken, so only direct costs were considered.
- Annual discounting of 3.5% was applied on costs and outcomes.
- The time horizon was 5 years.
- HbA1c progression for all arms was projected based on the progression in the EMPA-REG OUTCOME trial.
- The effects of the CVOTs were applied for the full 5 years, no treatment switch was foreseen.
- UK unit costs of complications and quality of life data were taken from literature. The drug costs were from the British National Formulary and Monthly Index of Medical Specialities with all the costs reported in 2015 British Pounds (GBP)
- The analysis included patients from EMPA-REG OUTCOME trial with mean age 63 years and 71% male with 9 years of duration of diabetes and 8.07% HbA1c (Table 2)
- The treatment effect on physiological parameters was applied in the CDM in two ways:
 - ✓ 1st year effect was programmed in the treatment settings together with the associated adverse event rates of each treatment (Table 3)
 - ✓ For 2nd and 3rd years, the treatment effect followed the progression over time available in respective trials of empagliflozin and liraglutide, while from 4th year onwards, HbA1c progression from the empagliflozin was applied for all competitors

Table 2. Patients baseline characteristics and risk factors (EMPA-REG OUTCOME trial, n=7020)

Characteristics	Mean (SE/SD)	Characteristics	Mean (SE/SD)
Patient Demographics			
Start age (years)	63.10 (8.6)	Prop. Male	0.71
Duration of Diabetes (years)	9		
Racial Characteristics			
Prop. White	0.543	Prop. Native American	0.008
Prop. Black	0.051	Prop. Asian/Pacific Islander	0.218
Prop. Hispanic	0.180		
Baseline Risk Factors			
HbA1c (%-points)	8.07 (0.85)	White Blood Cell (10^6/ml)	6.8
SBP (mmHg)	135.47 (17.0)	Heart rate (bpm)	68.47
DBP (mmHg)	76.67 (9.83)	Waist Hip Ratio (1 unit)	0.93
T-CHOL (mg/dL)	162.90 (43.80)	uAER* (mg/mmol)	19.3 (2.9)
HDL (mg/dL)	44.40 (11.70)	Serum Creatinine (mg/dl)	1.1
LDL (mg/dL)	85.60 (35.70)	Serum Albumin (g/dl)	3.9
TRIG (mg/dL)	170.50 (126.9)	Prop. smoker [0-1]	0.132
BMI (kg/m2)	30.62 (5.26)	Cigarettes/day**	3
eGFR (ml/min/1.73m2)	74.04 (21.41)	Alcohol consumption** (Oz/week)	3
Haemoglobin (gr/dl)	14.5		

* urinary albumin excretion rate **CDM Default value

Table 3. Absolute treatment effect on risk factors and adverse events within the CDM

Parameters	EMPAGLIFLOZIN[1,2]	LIRAGLUTIDE[3]
Glycated haemoglobin (HbA1c)*	-0.58	-1.37
Systolic Blood Pressure (SBP) (mmHg)*	-3.9	-1.82
Diastolic Blood Pressure (DBP) (mmHg)*	-1.72	0.17
Total cholesterol (T-Chol) (mg/dl)*	7.81	0
High-density lipoprotein (HDL) (mg/dl)*	1.81	0
Low-density lipoprotein (LDL) (mg/dl)*	4.79	0
Body mass index (BMI) (kg/m^2)*	-0.64	-0.88
Estimated glomerular filtration rate (eGFR) (ml/min/1.73m2)*	-0.16	0
Non-severe hypoglycaemia (NSHE) rate**	13.62	289.12
Severe hypoglycaemia (SHE1) rate**	0.44	8.24
Severe hypoglycaemia (SHE2) rate**	0.06	1.10

*Effect on the surrogate endpoints is applied on the first year of treatment; **Rate per 100 patient-year; SHE1: Severe hypoglycemia not needing any medical attention; SHE2: SH needing medical attention.

- Annual treatment cost includes drug cost and insulin cost plus the needle cost and self-monitoring of blood glucose (SMBG) where applicable.
- Annual cost of empagliflozin was £817 for 1st year and £834 for 2nd year onwards, while liraglutide has £1,742 and £1,945 for 1st year and for 2nd year onwards respectively.
- A probabilistic sensitivity analysis was also performed.

Results

Base case Results

- In the base case analysis, empagliflozin showed additional LE and higher QALY at a lower cost resulting in dominance of empagliflozin compared to liraglutide (Table 4).

Table 4. Cost-effectiveness results

	EMPAGLIFLOZIN	LIRAGLUTIDE
LE	4.254	4.201
QALY	2.789	2.685
Total costs (GBP)	13,973	18,432
ICUR (QALY)	Empagliflozin dominant	
ICER (LY)	Empagliflozin dominant	

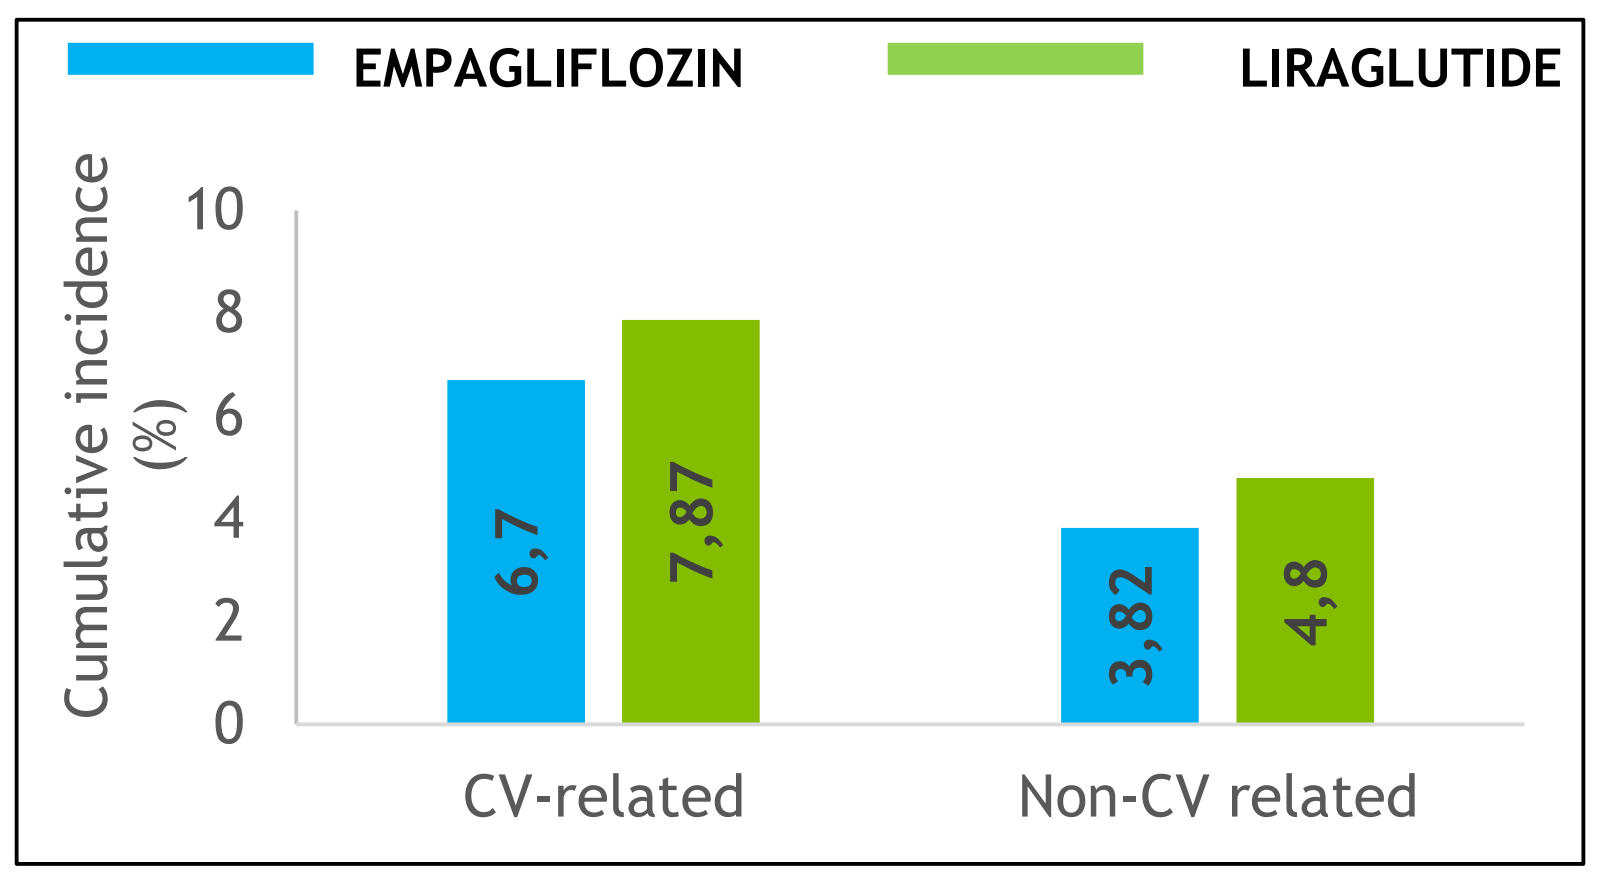
EMPAGLIFLOZIN
LIRAGLUTIDE
Cumulative incidence (%)
10
8
6
4
2
0
6,7
7,87
3,82
4,8
CV-related
Non-CV related

Figure 1 . 5-year cumulative incidence of Mortality

- At 5 years, empagliflozin has lower risk of CV and non-CV death as compared to liraglutide (Figure 1).
- Empagliflozin has a lower projected incidence of heart failure, but angina, stroke and PVD incidence were higher (Figure 2).
- Empagliflozin has lower total cost which was attributed to liraglutides higher treatment cost.
- Empagliflozin has higher cost of treating CVD complications, which was driven by the high cost of stroke management (Table 5).

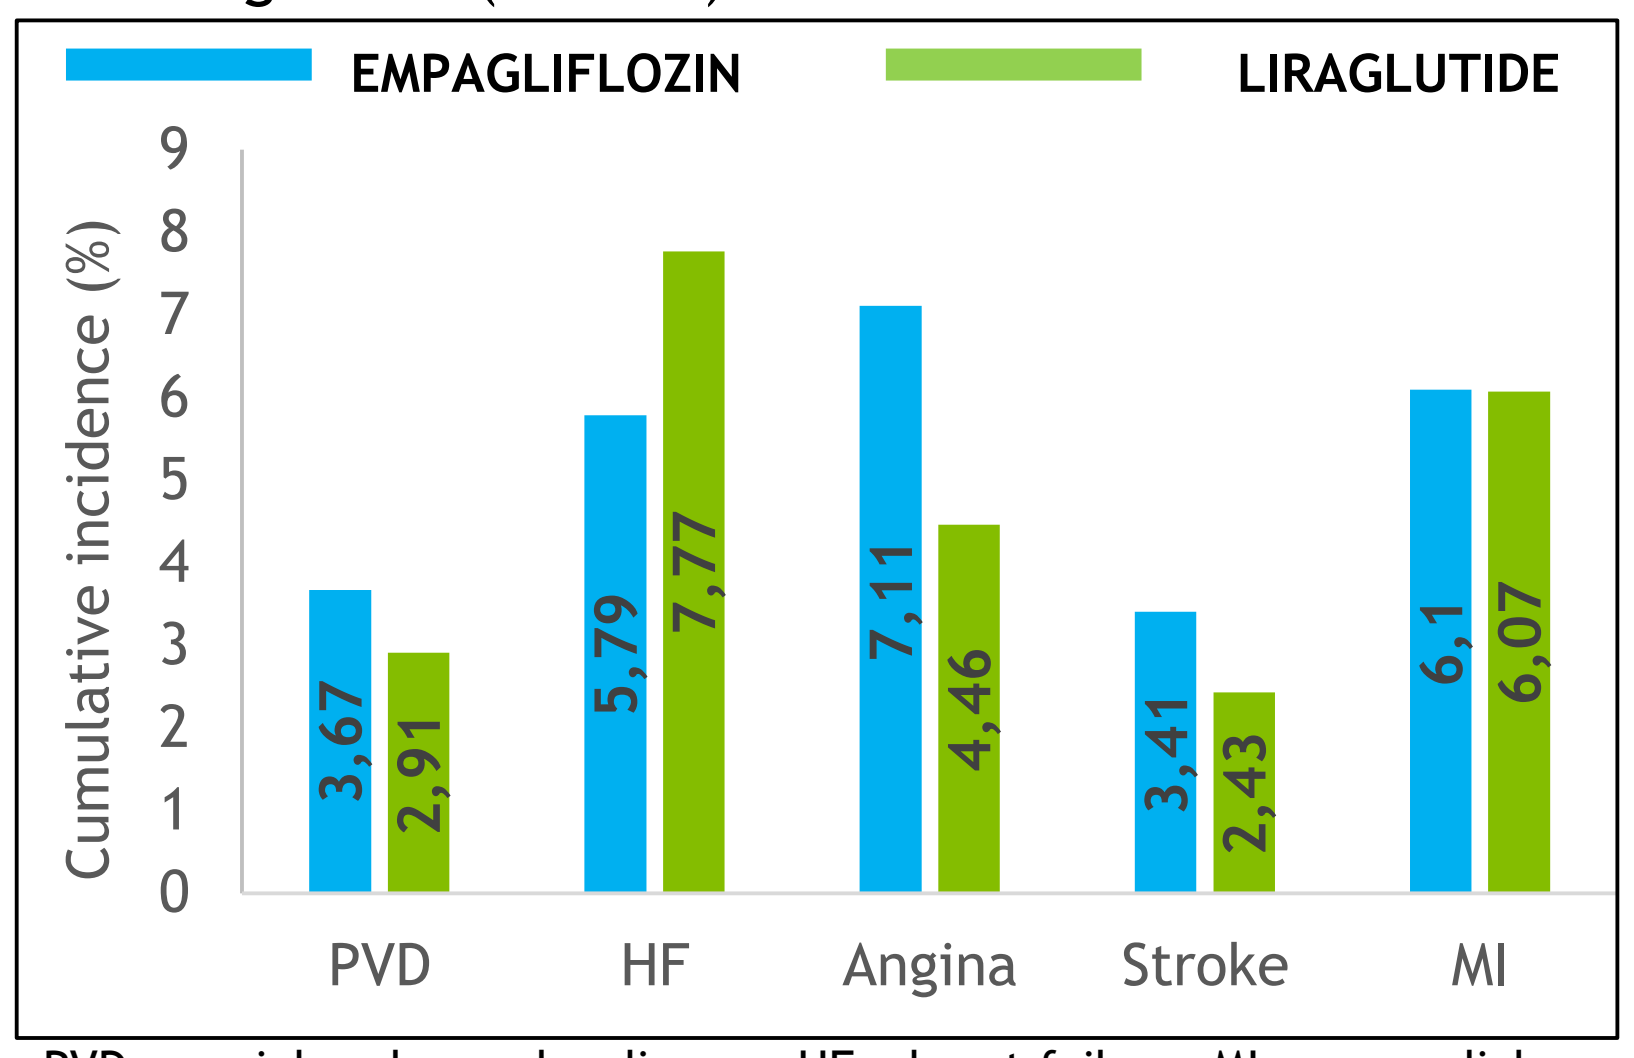
EMPAGLIFLOZIN
LIRAGLUTIDE
Cumulative incidence (%)
9
8
7
6
5
4
3
2
1
0
3,67
2,91
5,79
7,77
7,11
4,46
3,41
2,43
6,1
6,07
PVD
HF
Angina
Stroke
MI

PVD = peripheral vascular disease; HF = heart failure; MI: myocardial infarction

Figure 2. 5-year cumulative incidence of CVD complications

Table 5. Cost-breakdown per treatment arm (GBP)

	EMPAGLIFLOZIN	LIRAGLUTIDE
Treatment	3,610	8,196
Management	245	241
Cardiovascular disease	9,147	8,839
Kidney disease	576	538
Ulcer/Amputation/ Neuropathy	110	104
Eye disease	259	226
Hypoglycemia	28	288

Probabilistic Sensitivity Analysis

- The probabilistic sensitivity analyses performed on the base case demonstrated that Empagliflozin is cost-effective vs. liraglutide in 81% of the simulations for a willingness to pay threshold of £20,000 per QALY, in which dominance is only observed in 69% of the simulations reports (Figure 3 and 4).

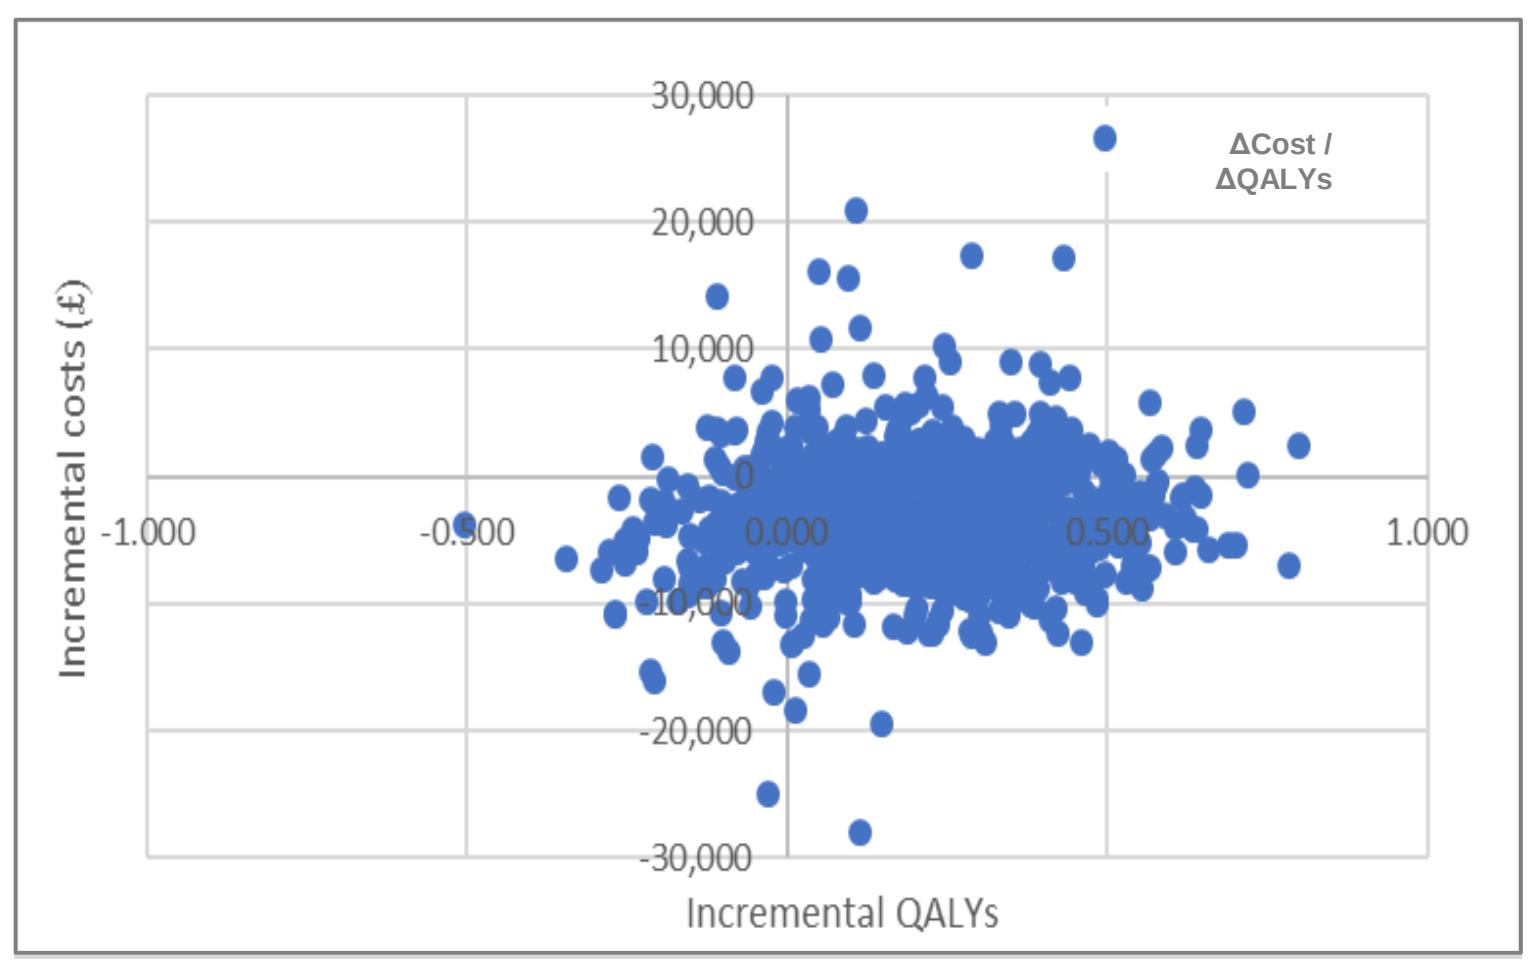
30,000
20,000
10,000
-20,000
-30,000
-1.000
-0.500
0.000
0.500
1.000
ΔCost / ΔQALYs
Incremental costs (£)
Incremental QALYs

Figure 3. Cost-effectiveness Scatter plot

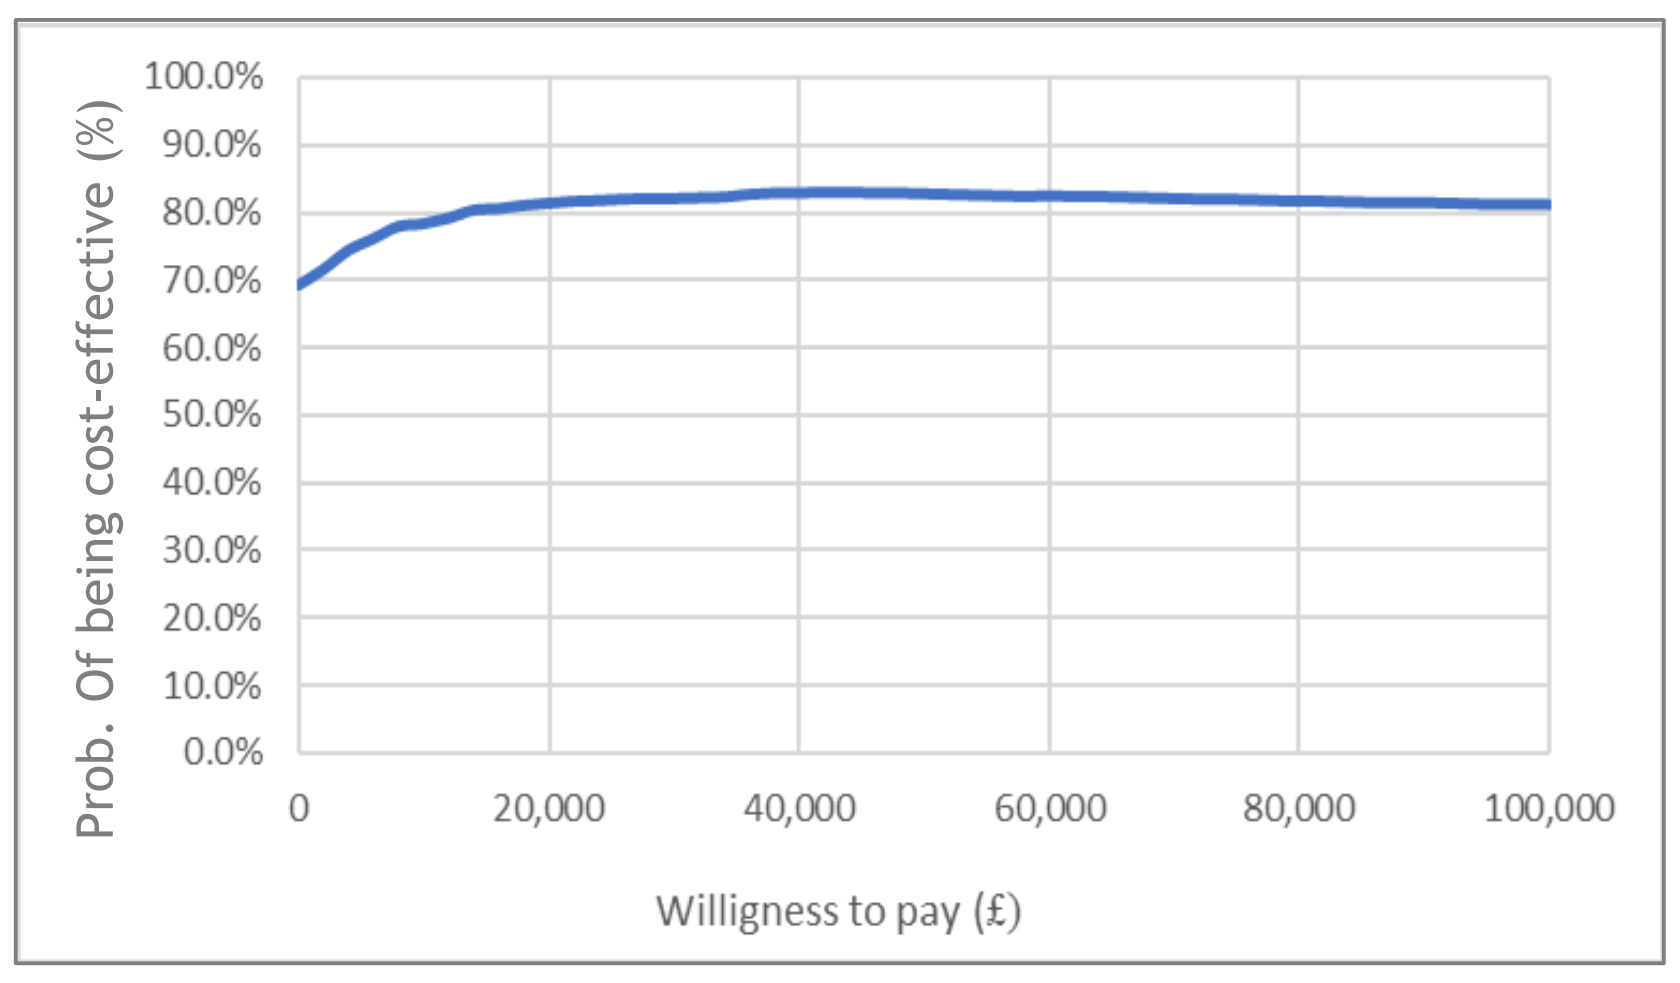
100.0%
90.0%
80.0%
70.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
0
20,000
40,000
60,000
80,000
100,000
Prob. Of being cost-effective (%)
Willingness to pay (£)

Figure 4. Cost-effectiveness Acceptability curve

Conclusion

The cost-effectiveness analyses based on the results of the EMPA-REG OUTCOME and LEADER trials, demonstrate that empagliflozin + SoC is dominant compared to Liraglutide + SoC from the UK NHS perspective.

References: 1. Zinman et al N Engl J Med. 2015;373(22):2117-2128. 2. Wanner et al N Engl J Med. 2016 Jul 28;375(4):323-34. 3. Marso et al N Engl J Med. 2016 Jul 28;375(4):311-22. 4. Balijepalli et al Diabetes Ther. 2018;9(4):1491-500.

Disclosure

IQVIA, the employer of MR, VF, and ML, received consulting fees from Boehringer Ingelheim to conduct the current work.

AU, NH and PG are/were full-time employees of Boehringer Ingelheim

ISPOR Europe 2018, Barcelona, Spain, 10–14 November 2018

Copyright © 2018 IQVIA. All rights reserved.