Treatment of atopic dermatitis in Portugal: insights from a physicians' survey

Carmo M[1], Faria M[2], Mendes-Bastos P[3]

1 – IQVIA, Oeiras, Portugal; 2 – Sanofi, Oeiras, Portugal; 3 – Dermatology Center, Hospital CUF Descobertas, Lisbon, Portugal

PSS57

ISPOR Europe 2018
10 – 14 November 2018
Barcelona, Spain

BACKGROUND AND OBJECTIVE

BACKGROUND

- Atopic Dermatitis (AD) is a chronic inflammatory skin disease estimated to affect up to 2 - 8% of adults in most countries of the world, with epidemiologic studies reporting variable prevalence across countries.
- To our knowledge, there are currently no guidelines for the treatment of AD in Portugal, and little is known about the standard of care provided to these patients, as well as about outcomes obtained with current treatment.

OBJECTIVE

- The study aimed at assessing physicians' prescribing behavior regarding the treatment of adults with Atopic Dermatitis (AD) in Portugal, as well as their perception on treatments and unmet needs.

METHODS

STUDY DESIGN

- A physician survey was conducted through computer assisted web interviewing (CAWI).
- Nine in-depth face-to-face interviews were also conducted with physicians to support survey design and to validate its outcomes.
- A total of 59 physicians were screened to reach the target sample of 50 physicians who responded to the complete survey, including 40 dermatologists and 10 allergists.

RESPONDENT SELECTION

- Physicians were randomly screened within the two specialties, to select those who would respond to the complete survey. The following selection criteria were used:
 - Physicians seeing ≥10 AD adult patient per month;
 - Physicians seeing ≥10 moderate AD adult patient per year;
 - Physicians seeing ≥2 severe AD adult patient per year.
- The sample included physicians treating AD patients in public sector, private sector or in both (66% of physicians stated treating patients both in public and private sector).

APPROACH

- Descriptive analysis was used to characterize the physicians' prescribing behavior and distribution of treated patients in the past 12 months, by type of treatment and response to treatment. Physicians were also asked to rank the importance of factors guiding their treatment choice.
- AD severity classification into mild, moderate or severe depends on individual physician's judgement. This option resulted from initial face-to-face interviews who revealed lack of viability in using standard scales classification due to lack of experience using them. Likewise, the classification of patients as "uncontrolled" is based on individual physician's judgement.
- Physicians were asked to indicate how many of their reported number of adult patients with moderate to severe AD had been treated in the past 12 months, and to distribute them according to a list of treatments.

FUNDING AND DISCLOSURES

- The study was funded by Sanofi and conducted by IQVIA.
- Carmo M is a IQVIA employee, Faria M is Sanofi employee. Mendes-Bastos P has received fees as consultant and/or speaker from AbbVie, Pfizer, Janssen-Cliag, Leo-Pharma, Novartis, Sanofi, Teva, Bayer and La Roche Posay and has worked as an Investigator in Clinical Trials sponsored by Abbvie and Novartis.

REFERENCES

1. *European Dermatology Forum (EDF). 2018. EDF Guidelines for Treatment of Atopic Eczema (Atopic Dermatitis). Developed by the Guideline Subcommittee "Atopic Eczema"*
2. *Eichenfield et al. 2014. Guidelines of care for the management of atopic dermatitis. American Academy of Dermatology (AAD). 2014 Feb;70(2):338-51*
3. *IQVIA, One Key*

RESULTS

- The study focused on moderate to severe AD adult patients followed either by dermatologists or allergists, which were estimated to represent 40% to 45% of adults with AD in 2017.
- Figure 1 shows the result of importance ranking of factors influencing treatment choice. For 50% of surveyed physicians, efficacy in reducing the eczema was the most important factor influencing prescription choice, followed by efficacy in reducing pruritus. Efficacy on pruritus was considered one of top 3 factors by 88%, whereas efficacy on eczema was in the top 3 for 84% of physicians.

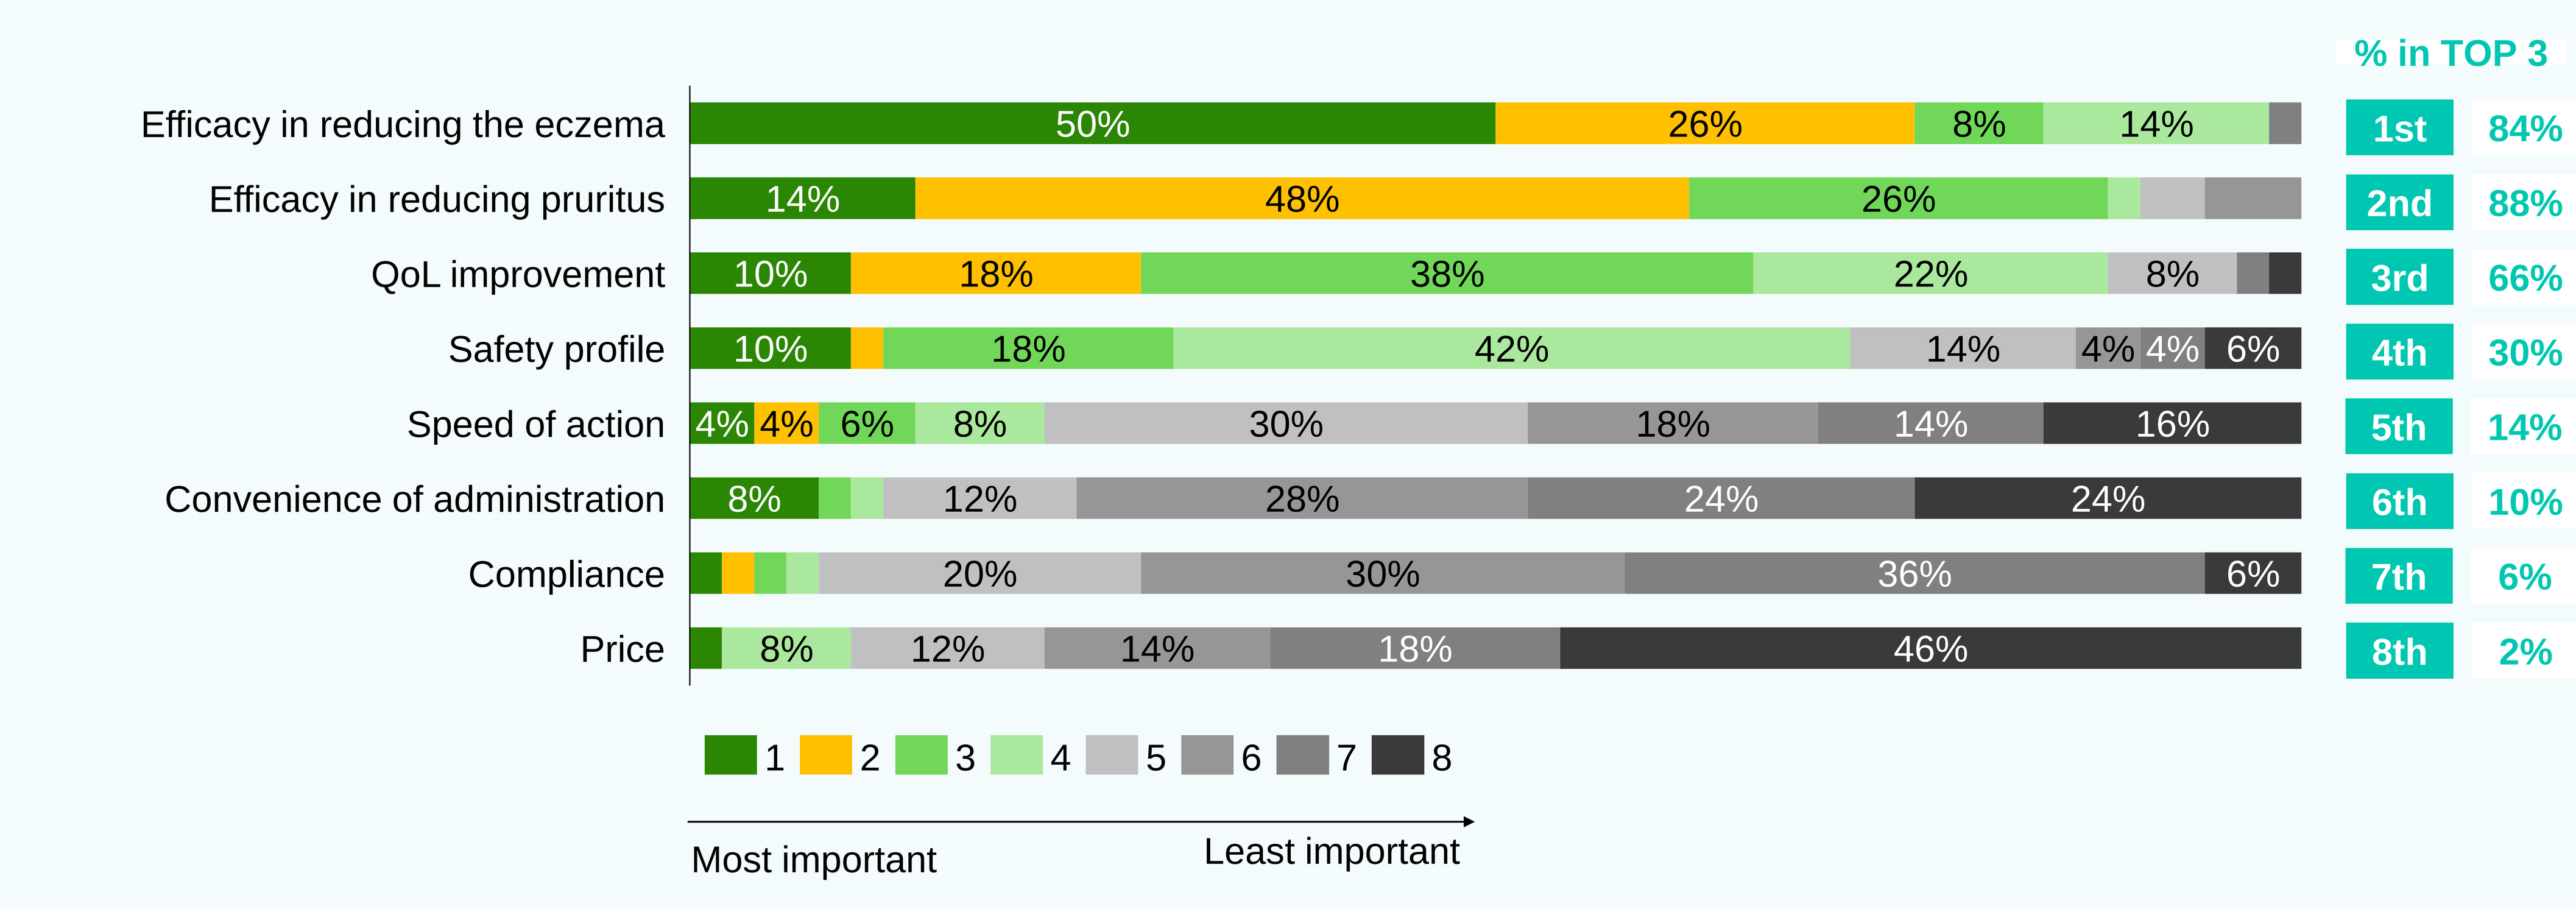
% in TOP 3
Efficacy in reducing the eczema
50% 26% 8% 14%
1st 84%
Efficacy in reducing pruritus
14% 48% 26%
2nd 88%
QoL improvement
10% 18% 38% 22% 8%
3rd 66%
Safety profile
10% 18% 42% 14% 4% 4% 6%
4th 30%
Speed of action
4% 4% 6% 8% 30% 18% 14% 16%
5th 14%
Convenience of administration
8% 12% 28% 24% 24%
6th 10%
Compliance
20% 30% 36% 6%
7th 6%
Price
8% 12% 14% 18% 46%
8th 2%
1 2 3 4 5 6 7 8
Most important
Least important

Figure 1. Importance ranking of factors influencing treatment choice by physicians
(% of physicians selecting the attribute by order of importance)

- Physicians were then asked to classify each treatment option on a scale of 1 to 10 according to each of these factors. As illustrated in Figure 2, physicians' classification of treatment options simultaneously positioned those regarded as most effective as being less safe – namely oral corticosteroids (OCS), cyclosporine and other immunosuppressants).

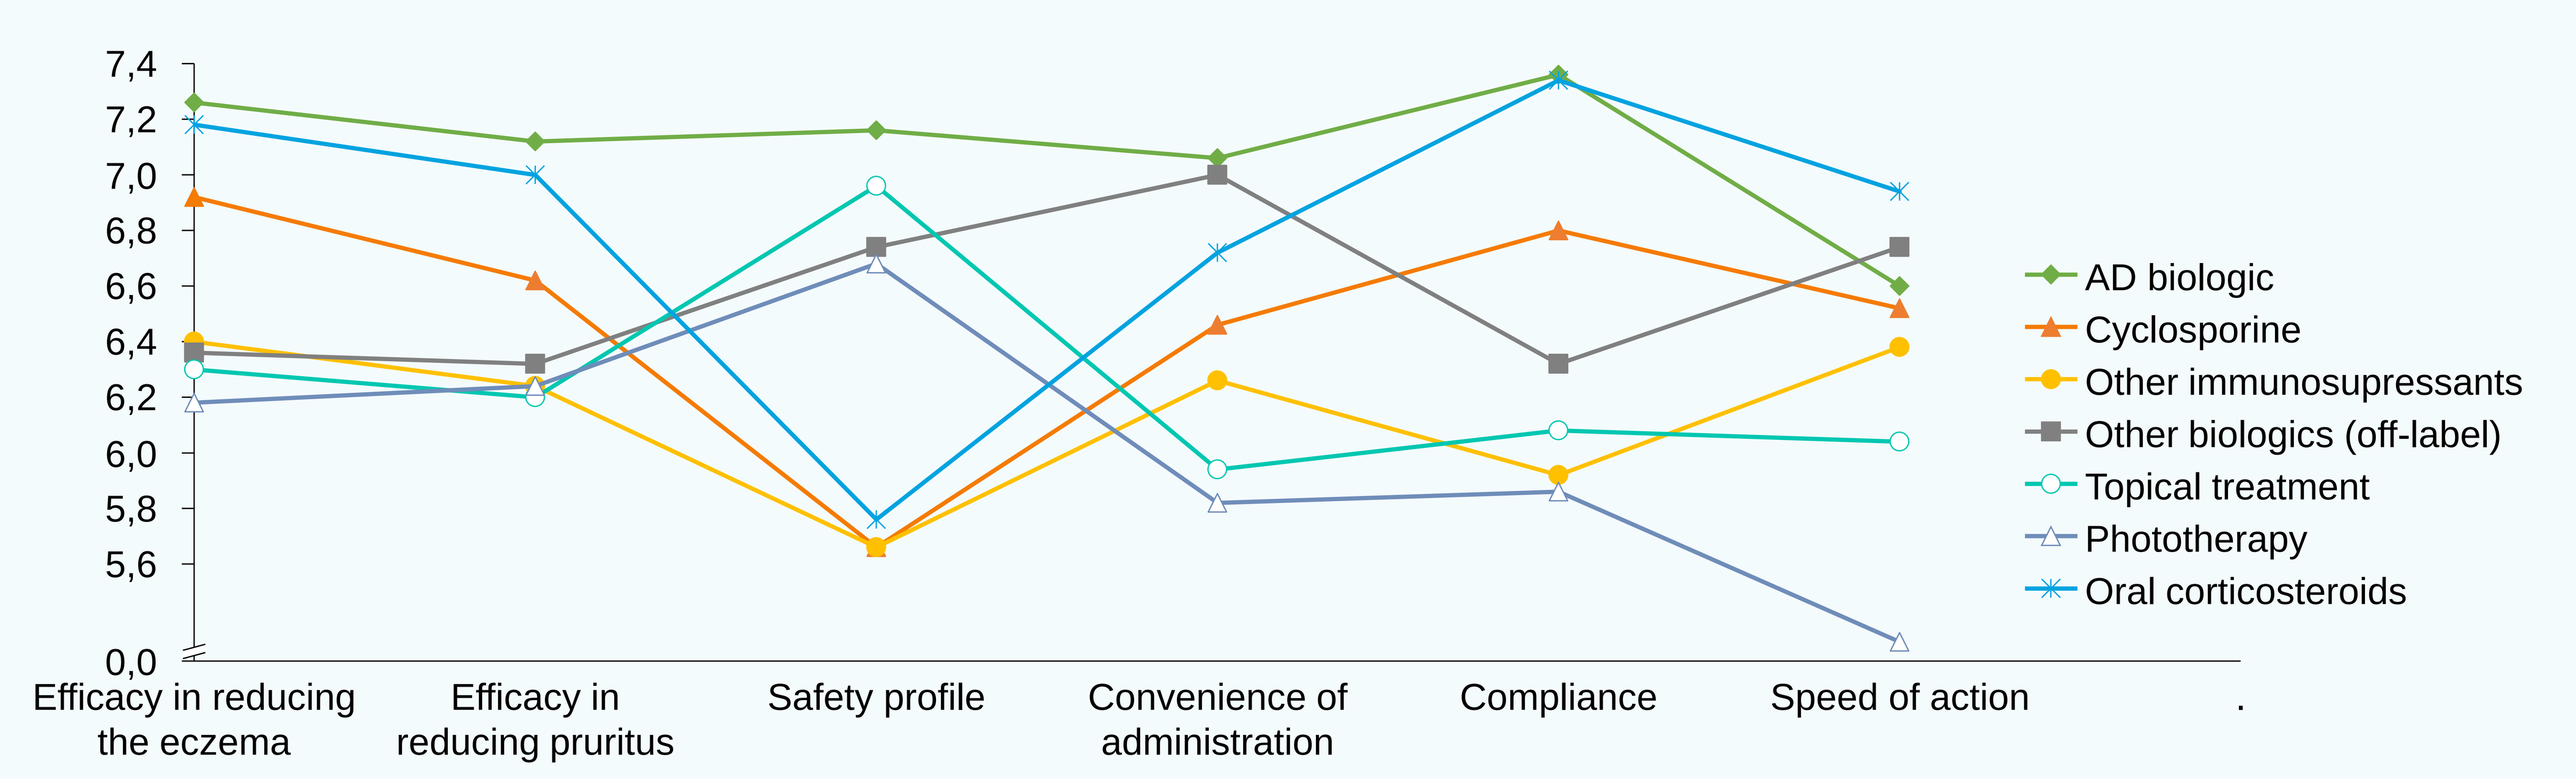
7,4
7,2
7,0
6,8
6,6
6,4
6,2
6,0
5,8
5,6
0,0
Efficacy in reducing the eczema
Efficacy in reducing pruritus
Safety profile
Convenience of administration
Compliance
Speed of action
AD biologic
Cyclosporine
Other immunosupressants
Other biologics (off-label)
Topical treatment
Phototherapy
Oral corticosteroids

Figure 2. Physician's grading of AD treatment's performance
(average doctors classification on a scale of 1 to 10, where 10 represents the highest performance possible and 1 the lowest)

- According to surveyed physicians, only 91.4% of their moderate to severe AD adult patients were treated in the past 12 months. Within the 8.6% of untreated patients, 35.7% were untreated due to contraindications, 35.7% due to previous lack of response, and 28.6% due to patient decision.
- Amongst treated patients, 37.5% were exclusively on topical treatment - including topical corticosteroids (TCS) and topical calcineurin inhibitors (TCI) – while the remaining received other treatments (Figure 3).
- Physicians estimate that 39.2% of their moderate to severe AD patients that were exclusively on TCS and/ or TCI over the past 12 months were uncontrolled with this therapeutic, representing approximately 13.4% of total patients (Figure 3).
- A total of 24.3% of moderate to severe patients were considered by their physicians as being uncontrolled with current treatment, of which 73.9% were on topical or OCS treatment.

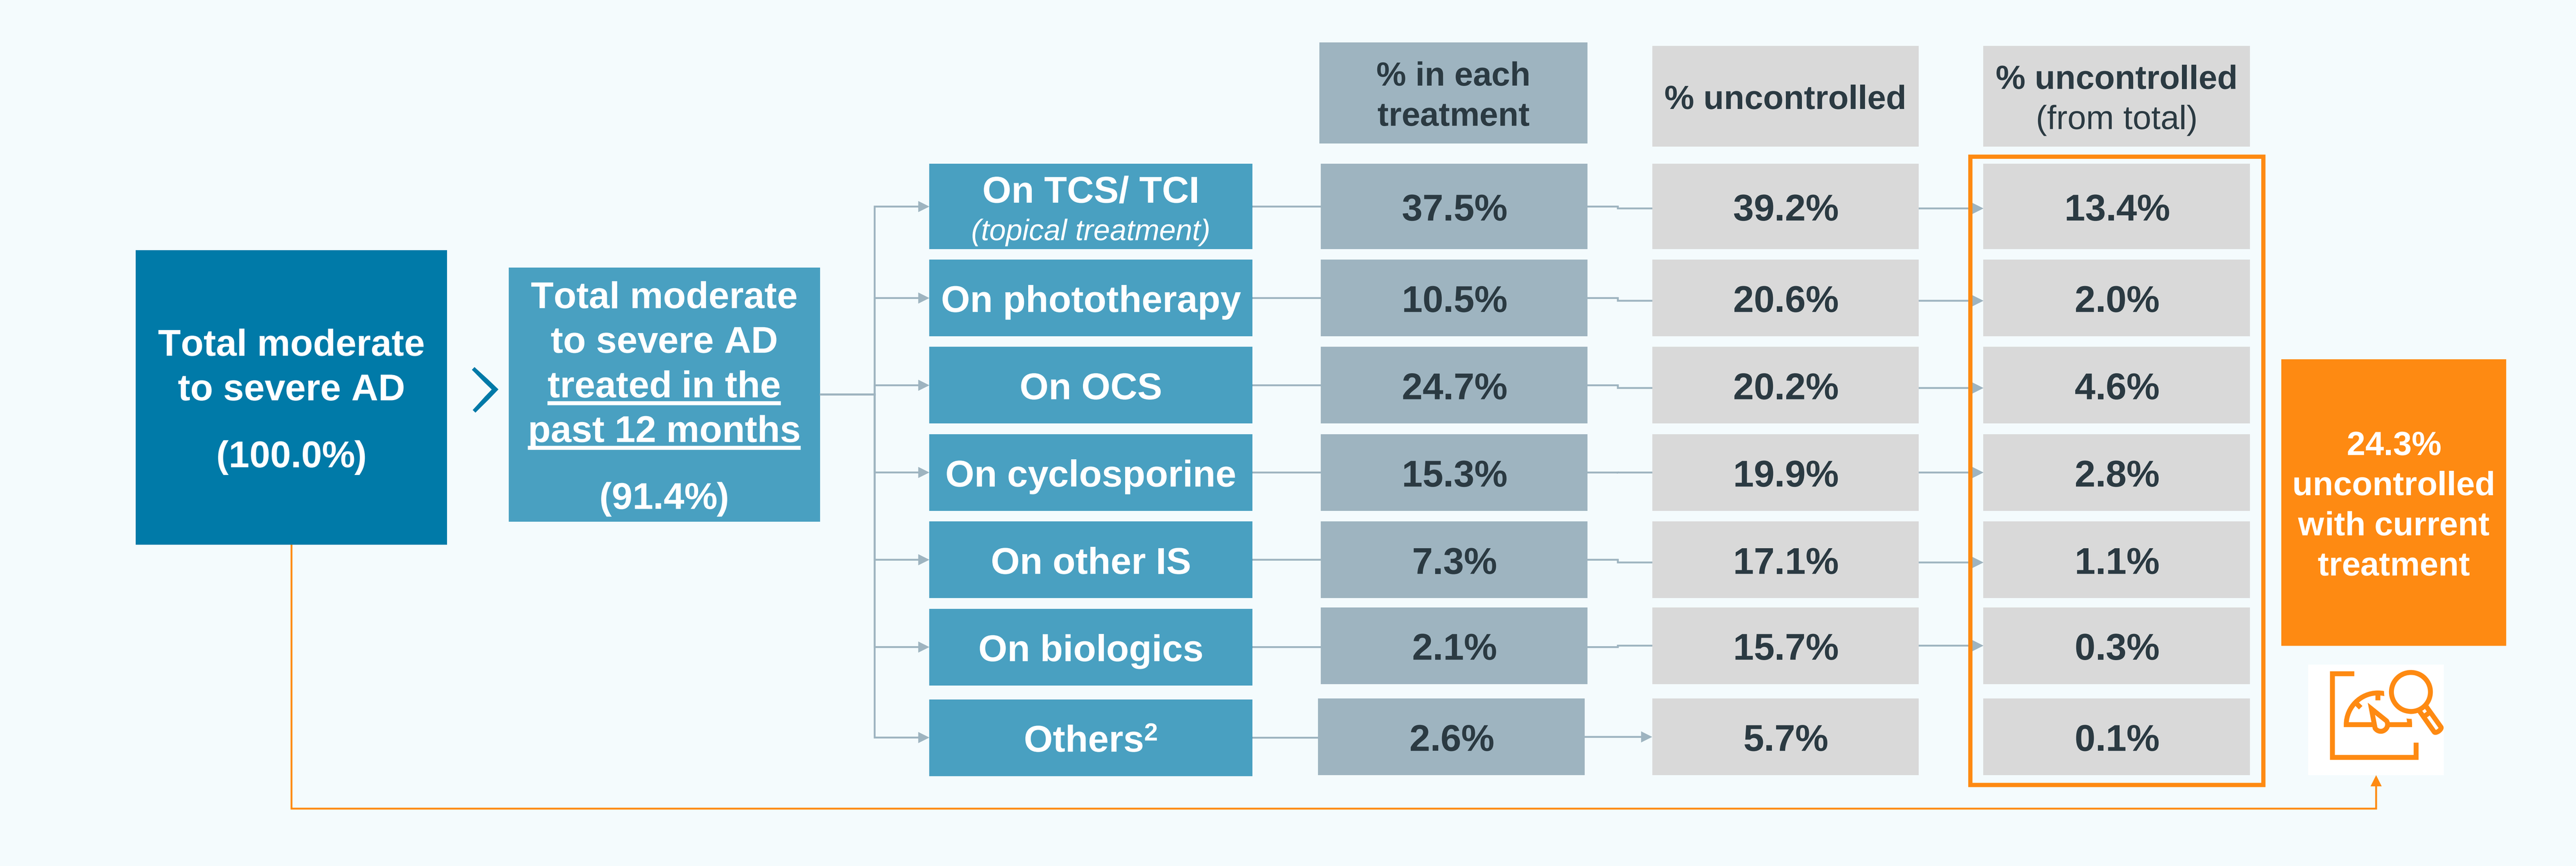
Total moderate to severe AD
(100.0%)
Total moderate to severe AD treated in the past 12 months
(91.4%)
% in each treatment
% uncontrolled
% uncontrolled (from total)
On TCS/ TCI (topical treatment) 37.5% 39.2% 13.4%
On phototherapy 10.5% 20.6% 2.0%
On OCS 24.7% 20.2% 4.6%
On cyclosporine 15.3% 19.9% 2.8%
On other IS 7.3% 17.1% 1.1%
On biologics 2.1% 15.7% 0.3%
Others² 2.6% 5.7% 0.1%
24.3% uncontrolled with current treatment

Figure 3. Distribution of treated patients in the past 12 months, by type of treatment and response to treatment
(% of patients)

CONCLUSIONS

- Based on physicians' surveys and in-depth interviews, a comprehensive understanding of AD treatment prescribing behavior was attained.
- One-third of moderate to severe AD adult patients were either uncontrolled with their current treatment (24%) or untreated (9%), suggesting a high unmet need.
- The worst grading regarding cyclosporine, OCS and other immunosuppressants was the safety profile, emphasizing the need for safe options for long term AD management.