Budget impact analysis of liposomal amphotericin B (L-AmB) versus amphotericin B lipid complex (ABLC) for the treatment of invasive fungal infections in a tertiary hospital in the Kingdom of Saudi Arabia

Al Jedai A[1], AlRaddadi B[2], Vemer P[3], Awad N[4], Safwat M[4], Mansour E[5], Hanna E[5]

[1]King Faisal Specialist Hospital and Research Centre, Riyadh, Saudi Arabia, [2]King Faisal Specialist Hospital, Jeddah, Saudi Arabia, [3]Groningen University, Groningen, The Netherlands, [4]IQVIA, Dubai, United Arab Emirates, [5]Gilead, Dubai, United Arab Emirates

IMS Health & Quintiles are now IQVIA™

INTRODUCTION

- Invasive fungal infections (IFIs) are an emerging problem worldwide, largely due to increased use of immunosuppressive agents among certain populations (e.g. cancer and transplant patients). Almost 87% of all IFIs are caused by Candida and Aspergillus species.[1,2]
- Major risk factors for IFIs include neutropenia, haematological malignancies, bone marrow transplantation, prolonged corticosteroid therapy, prolonged stays in intensive care, chemotherapy, HIV infection, invasive medical procedures, and use of immune suppressive agents, (like Alemtuzumab, Daclizumab, etc).[3,4]
- Recent global estimates suggest that IFIs are responsible for 1.5 million deaths annually and have a mortality rate of around 40% worldwide.[5] In the United States, the clinical and economic burden of IFIs was reported to be substantial, especially in transplant recipients. Patients with an IFI had, on an average, a 5-fold increase in mortality, an additional 19.2 hospital days, and $55,400 in excess costs compared with patients without an IFI.[6]
- High mortality rates were also reported from Kingdom of Saudi Arabia (KSA) ranging from 43 to 59%[6]. In the year 2012, incidence of IFIs was reported to be 1.95/1000 discharges in KSA.[7]
- Treatment options for IFIs globally include flucytosine, polyenes (amphotericin), triazoles (fluconazole, itraconazole, posaconazole, voriconazole) and echinocandins (anidulafungin, caspofungin , micafungin).[8]
- Amphotericin B (AmB) is commonly used for treatment of IFIs. Amphotericin B lipid complex (ABLC) (which is a 1:1 combination of AmB and lipid moiety) is indicated for the treatment of IFIs in patients who are intolerant of conventional AmB therapy, while Liposomal Amphotericin B (L-AmB) is indicated in patients with renal impairment or untolerated toxicity of AmB therapy.[9,10]
- L-AmB is a unique liposomal formulation of AmB. It is a single bilayer liposomal drug delivery system and has prolonged mean residence time in body making it efficacious even at lower doses. L-AmB in comparison to ABLC has shown better safety profile, tolerability profile and has lower haematological toxicity.[11]

OBJECTIVE

- This study evaluated the budgetary impact of introducing L-AmB versus ABLC (per year) for the treatment of IFIs in King Faisal Specialist Hospital and Research Centre (KFSHRC) in KSA

METHODOLOGY

- A Budget Impact Model (BIM) was developed to understand the economic effect of introduction of L-AmB in KSA.
- Model's considerations;
 - Model structure and perspective: Microsoft® Excel based model was developed from payer perspective.
 - Model Inputs: Various inputs such as estimated number of patients at KFSHRC treated for IFIs, direct costs based on drug dosing as well as drug acquisition, administration and adverse events management costs which were sourced from literature review and experts' interviews.
 - Model Outputs: The model assessed the change in the hospital budget following the introduction of L-AmB as treatment option in addition to ABLC. These were calculated in the form of two scenarios:
 - Scenario 1 – Current scenario with ABLC only
 - Scenario 2 – Anticipated scenario with L-AmB replacing ABLC as choice of therapy by 100%
 - Sensitivity Analysis: A one-way sensitivity analysis was performed to understand the impact on budget when various model inputs were increased or decreased by 20%.

Step 1
Understanding KSA Market for IFIs

Step 2
Model Concept Validation

Step 3
Model Devlopment

Step 4
Experts' Interveiws

Step 5
Model population with KSA specific data

Results and Manuscript

The Budget Impact Model (BIM) calculates the outcomes based on inputs such as

Current Market Share | **Anticipated Market Share** | **Patient Characteristics** | **Drug acquisition, administration & wasted vial cost** | **Adverse Events**

Outcomes were calculated for both current and anticipated market scenarios from a payer perspective

OUTCOMES

Sensitivity Analysis

RESULTS

- Introduction of L-AmB at KFSHRC demonstrated cost savings compared to ABLC with total savings of 22.8% (Fig 1). The reduction in total costs for L-AmB was driven by both lower medication costs and lower adverse event management costs.
- Use of L-AmB reduced medication cost by 20.5% while the cost of managing adverse events were 52.7% lower compared to ABLC. L-AmB decreased total medication cost compared to ABCL, by reducing the drug acquisition costs by 27.4%, drug administration cost by 2.9% and decreasing the vial wastage by 54.4% due to better dosing regimen (Fig 2).
- Adverse events with L-AmB were considerably lower than those with ABLC (Fig 3). Use of L-AmB lead to 59.6% decrease in nephrotoxicity compared to ABLC, while the relative difference for dialysis, infusion related reactions and hypomagnesaemia ranged between -36.6% and -60.1%.
- The sensitivity analysis (Fig 4) showed that increasing or decreasing the treatment duration and the AE rates by 20% for ABLC and L-AmB had the highest impact on both medication costs and total costs, compared to baseline.

RESULTS (contd.)

Fig. 1: Overall cost: *scenario 1 (ABLC) vs. scenario 2 (L-AmB)*

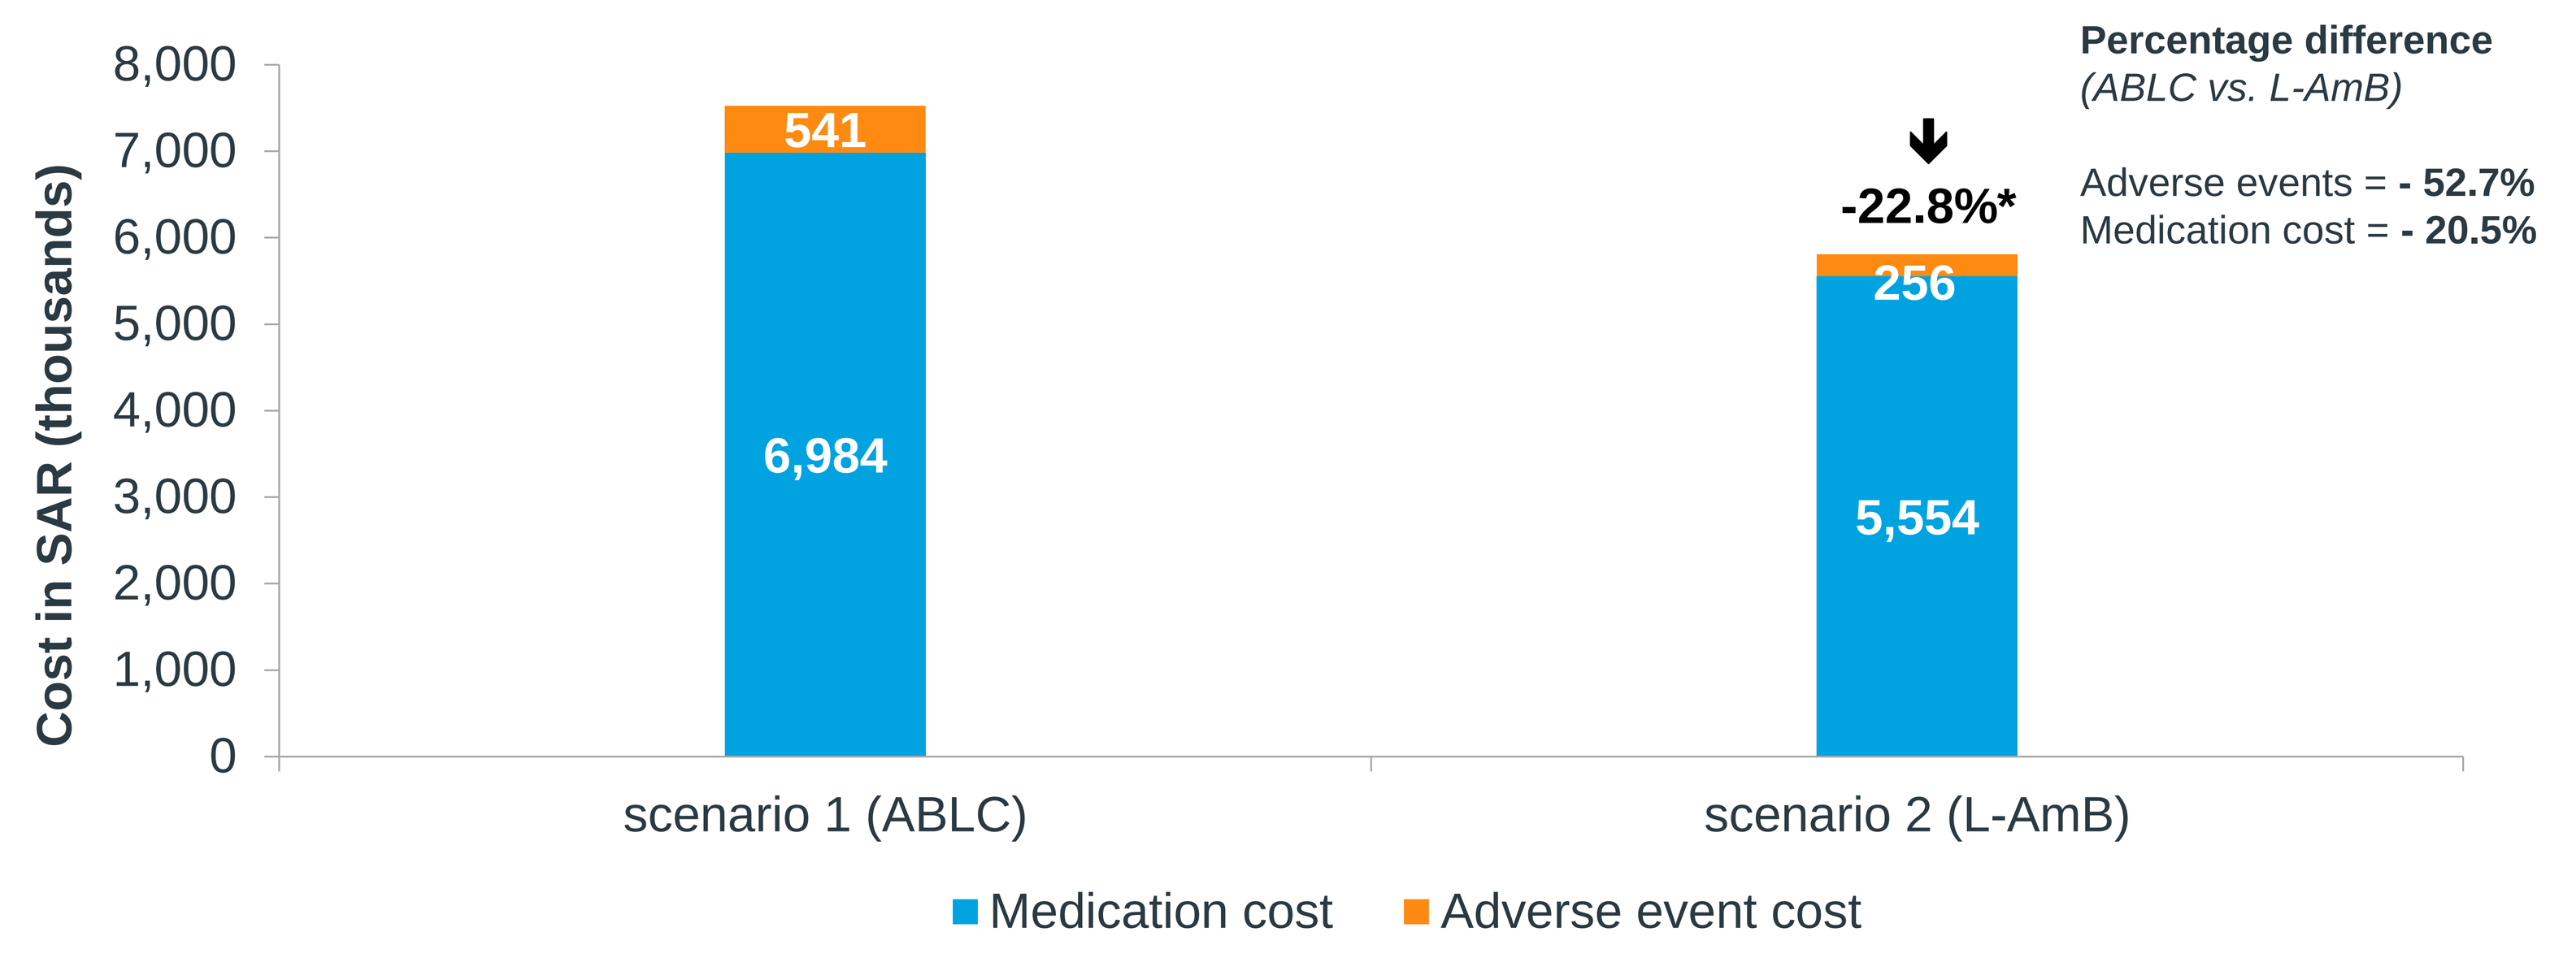

Cost in SAR (thousands)
8,000
7,000
6,000
5,000
4,000
3,000
2,000
1,000
0
541
6,984
256
5,554
-22.8%*
scenario 1 (ABLC)
scenario 2 (L-AmB)
Medication cost
Adverse event cost
Percentage difference
(ABLC vs. L-AmB)
Adverse events = - 52.7%
Medication cost = - 20.5%

*Percentage difference between scenario 1 & 2

Fig. 2: Medication cost: *scenario 1 (ABLC) vs. scenario 2 (L-AmB)*

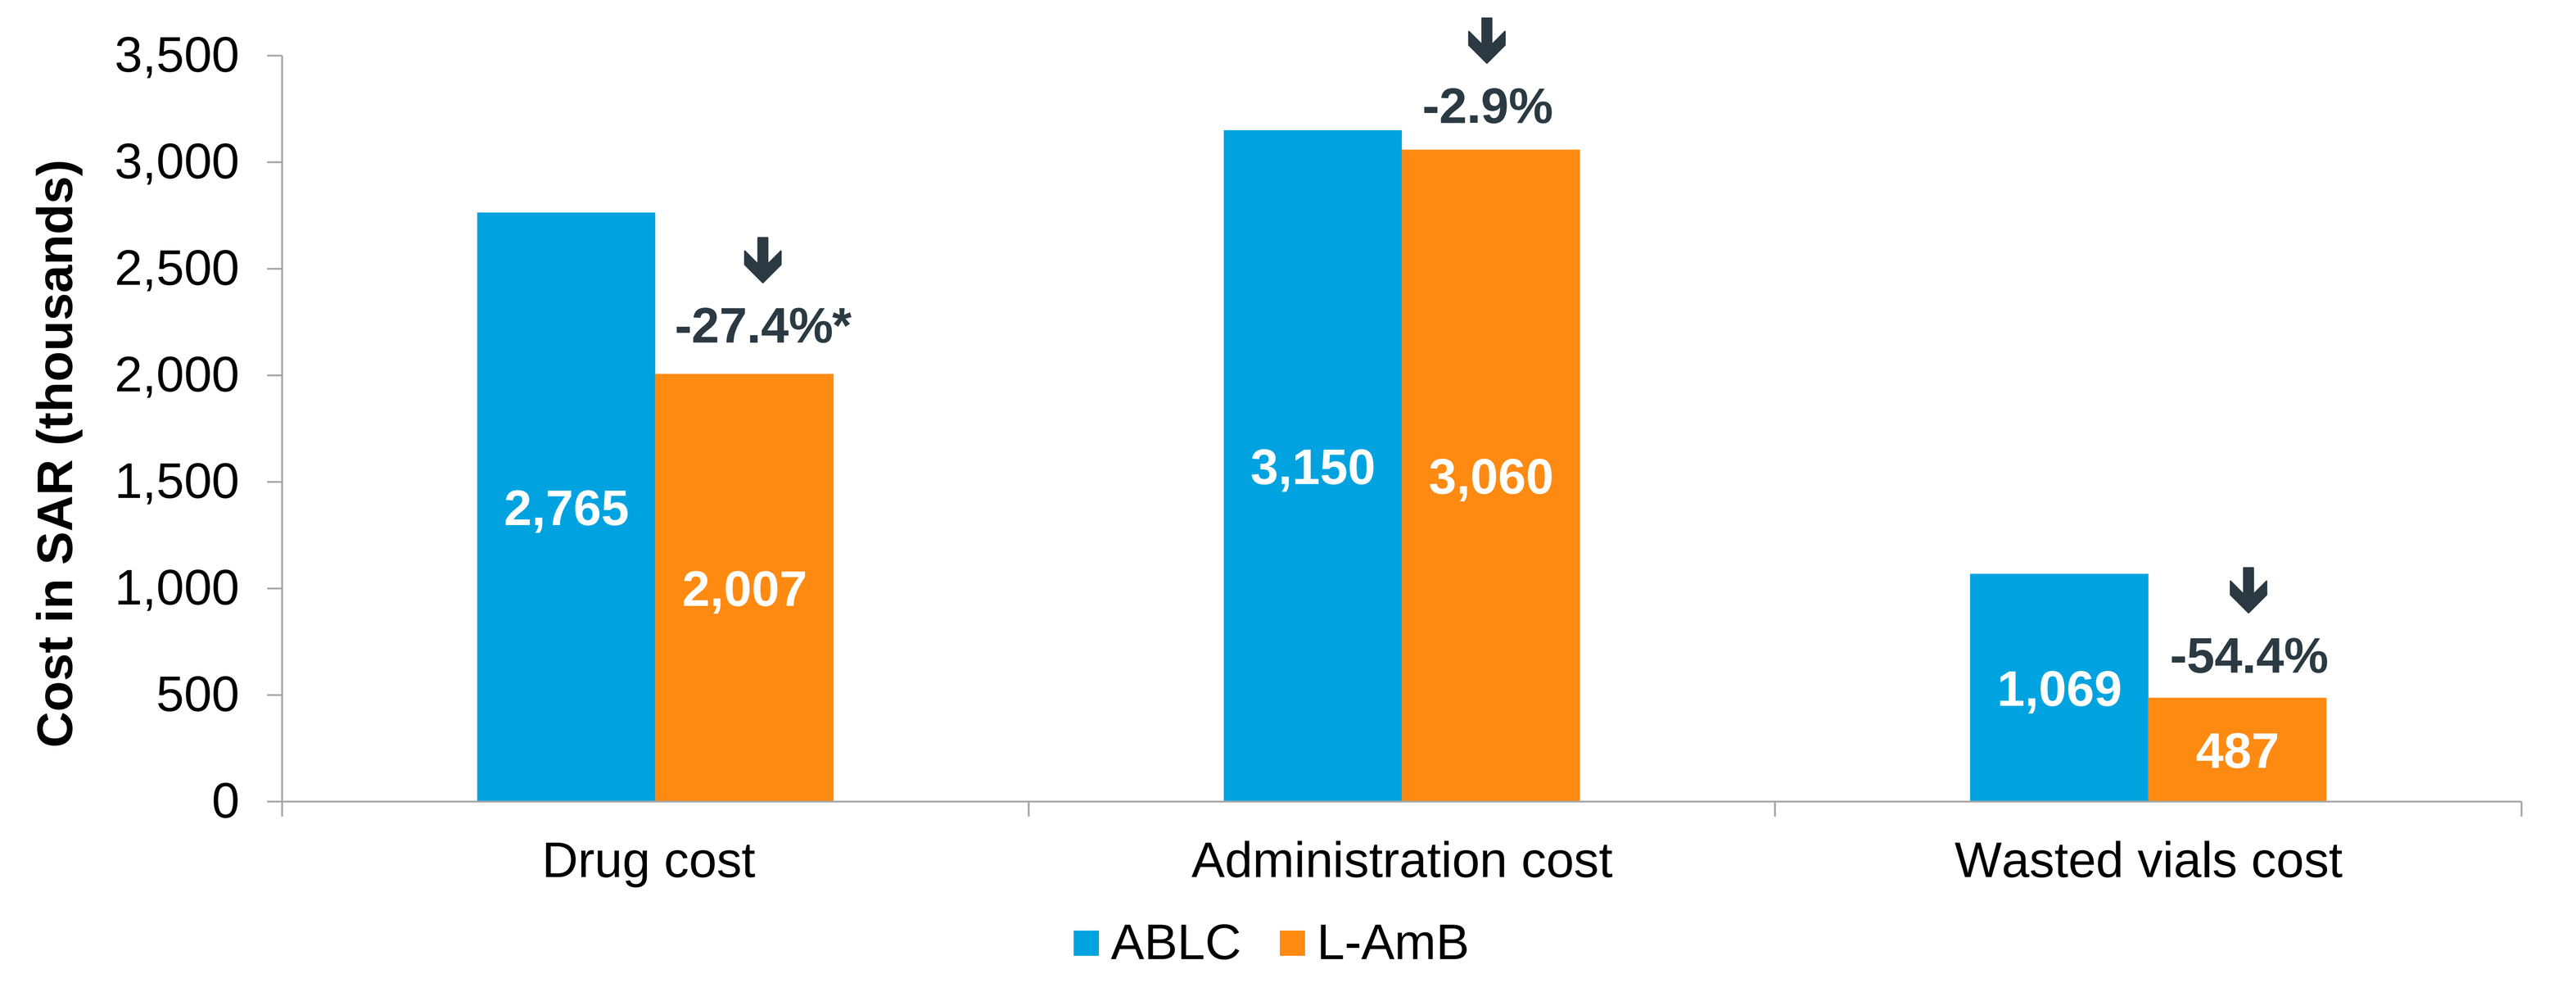

Cost in SAR (thousands)
3,500
3,000
2,500
2,000
1,500
1,000
500
0
2,765
2,007
-27.4%*
3,150
3,060
-2.9%
1,069
487
-54.4%
Drug cost
Administration cost
Wasted vials cost
ABLC
L-AmB

*Percentage difference between scenario 1 & 2

Fig. 3: Number of adverse events: *scenario 1 (ABLC) vs. scenario 2 (L-AmB)*

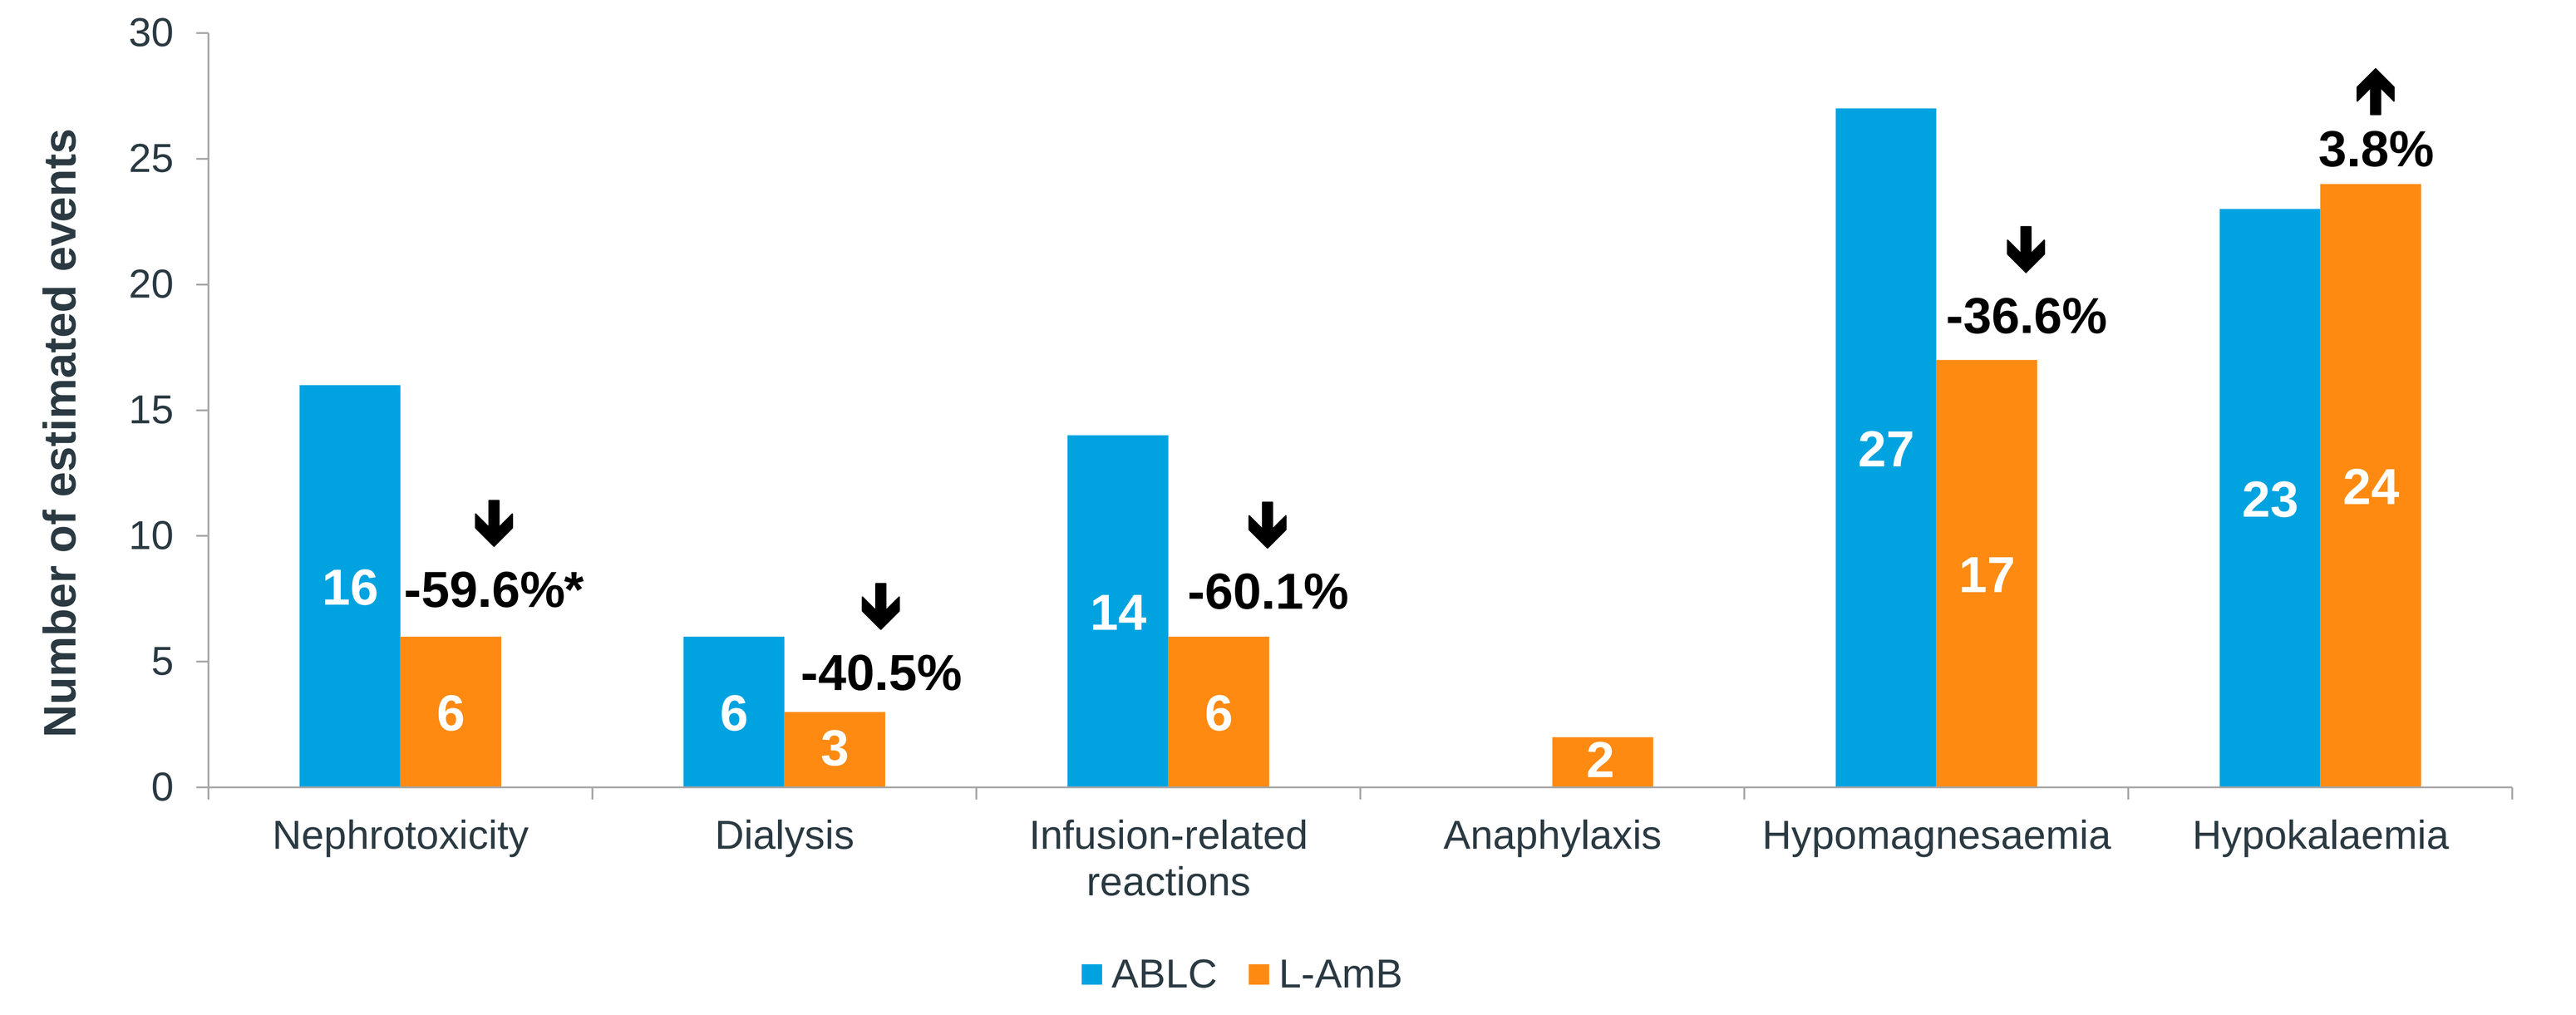

Number of estimated events
30
25
20
15
10
5
0
16
6
-59.6%*
6
3
-40.5%
14
6
-60.1%
2
27
17
-36.6%
23
24
3.8%
Nephrotoxicity
Dialysis
Infusion-related reactions
Anaphylaxis
Hypomagnesaemia
Hypokalaemia
ABLC
L-AmB

*Percentage difference between scenario 1 & 2

Fig. 4: One-way sensitivity analysis: *Total costs*

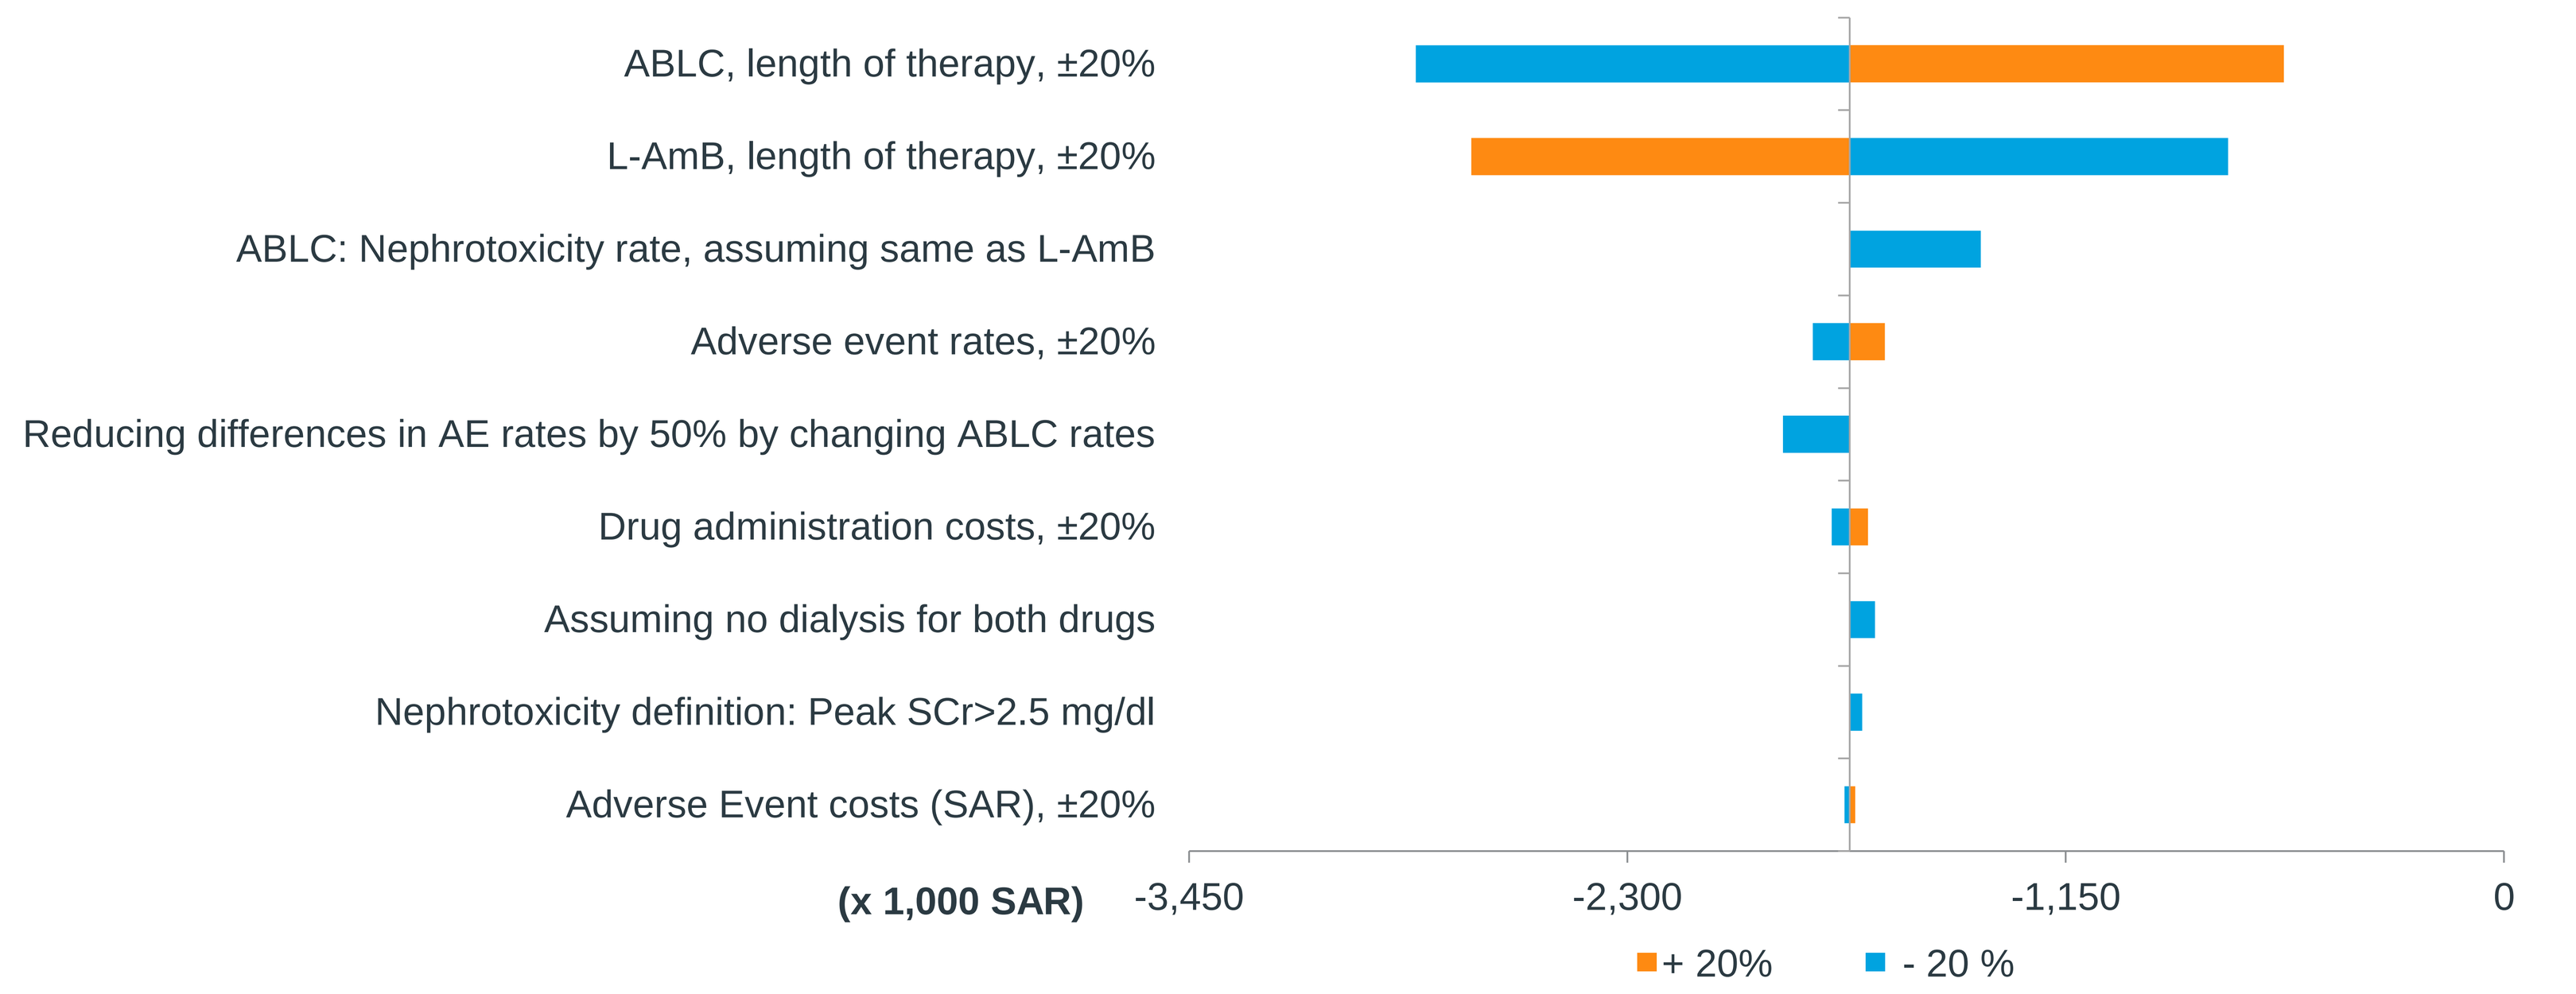

ABLC, length of therapy, ±20%
L-AmB, length of therapy, ±20%
ABLC: Nephrotoxicity rate, assuming same as L-AmB
Adverse event rates, ±20%
Reducing differences in AE rates by 50% by changing ABLC rates
Drug administration costs, ±20%
Assuming no dialysis for both drugs
Nephrotoxicity definition: Peak SCr>2.5 mg/dl
Adverse Event costs (SAR), ±20%
(x 1,000 SAR)
-3,450
-2,300
-1,150
0
+ 20%
- 20 %

*Inputs having low impact are dialysis cost, hypomagnesaemia, and infusion related cost

CONCLUSION

- The introduction of L-AmB in the KSA market is expected to be associated with more cost savings as compared to ABLC in management of IFIs.
- The above results are expected to support in communicating the value of the new intervention, which can be critical in informing pricing decisions, supporting discussions with different payers as well as reimbursement and formulary listings decision making processes.

References:

1. Gavalda, J., et al. (2014) Invasive fungal infections in solid organ transplant recipients. Clin Microbiol Infect, 20, pp.27-48
2. Paramythiotou, E., et al. (2014) Invasive fungal infections in the ICU: how to approach, how to treat. Molecules, 19(1), pp.1085-1119
3. Enoch, D.A., et al. (2006) Invasive fungal infections: a review of epidemiology and management options. J Med Microbiol, 55(7), pp.809-818
4. Maschmeyer, G. et.al. (2009) New immunosuppressive agents and risk for invasive fungal infections. Current infectious disease reports, 11(6), p.435
5. Bongomin, F., et al. (2017) Global and multi-national prevalence of fungal diseases—estimate precision. J Fungi, 3(4), p.57
6. Lass-Flörl C. (2009) The changing face of epidemiology of invasive fungal disease in Europe. Mycoses, 52, pp.197-205
7. Moghnieh, R., et.al (2017) Epidemiology and outcome of invasive fungal infections and methicillin-resistant Staphylococcus aureus (MRSA) pneumonia and complicated skin and soft tissue infections (cSSTI) in Lebanon and Saudi Arabia. J Infect Public Health, 10(6), pp.849-854
8. Pound, M.W., et. al..)2011) Overview of treatment options for invasive fungal infections. Med. Mycol, 49(6), pp.561-580
9. AmBisome® (amphotericin B) liposome for injection: USFDA Product Label (Assessed on 3th October 2018)
10. ABELCET® (Amphotericin B Lipid Complex Injection): USFDA Product Label (Assessed on 3th October 2018)
11. Stone, N.R., et.al. (2016). Liposomal amphotericin B (AmBisome®): a review of the pharmacokinetics, pharmacodynamics, clinical experience and future directions. Drugs, 76(4), pp.485-500

Copyright © 2018 IQVIA. All rights reserved.